Language impairment in progressive supranuclear palsy and corticobasal syndrome

Katie A. Peterson[1], Nikil Patel[2], Tom Barrick[2], Stefano F. Cappa[3], Ajay Halai[4], Ruth Ingram[4], Matthew A. Lambon Ralph[4], Karalyn E. Patterson[1], James B. Rowe[1], Peter Garrard[2]

[1]Department of Clinical Neurosciences, University of Cambridge, [2]Neuroscience Research Centre, St George's, University of London, [3]NeTS, Scuola Universitaria Superiore IUSS, Pavia, [4]Division of Neuroscience and Experimental Psychology, School of Biological Sciences, University of Manchester

Introduction

- Cognitive impairments are common in progressive supranuclear palsy (PSP) and corticobasal syndrome (CBS), however these symptoms are often overshadowed by the motor symptoms
- Language impairment is a common early symptom in patients with PSP and CBS, therefore, language assessment may have significant utility in their differential diagnosis
- Here, we present preliminary findings using the newly developed Mini Linguistic State Examination (MLSE) to examine the status of language in patients with PSP and CBS

Methods

Participants

Participants were patients with a diagnosis of probable PSP or CBS, primary progressive aphasia (PPA), and age-matched healthy controls

Table 1. Participant demographics

Group	N	Age M (SD)	M/F
PSP	10	69 (5.9)	7/3
CBS	9	68 (8.9)	4/5
Control	30	64 (5.7)	15/15
PPA	10	70 (6.4)	3/7

Procedure

Participants completed the Mini Linguistic State Examination (MLSE; see below), a language assessment tool featuring tasks which cover the range of domains affected in progressive aphasias (e.g. naming, word/sentence comprehension, semantic association, repetition, reading, and writing). Example test items are shown below

The Mini Linguistic State Examination (MLSE): example items

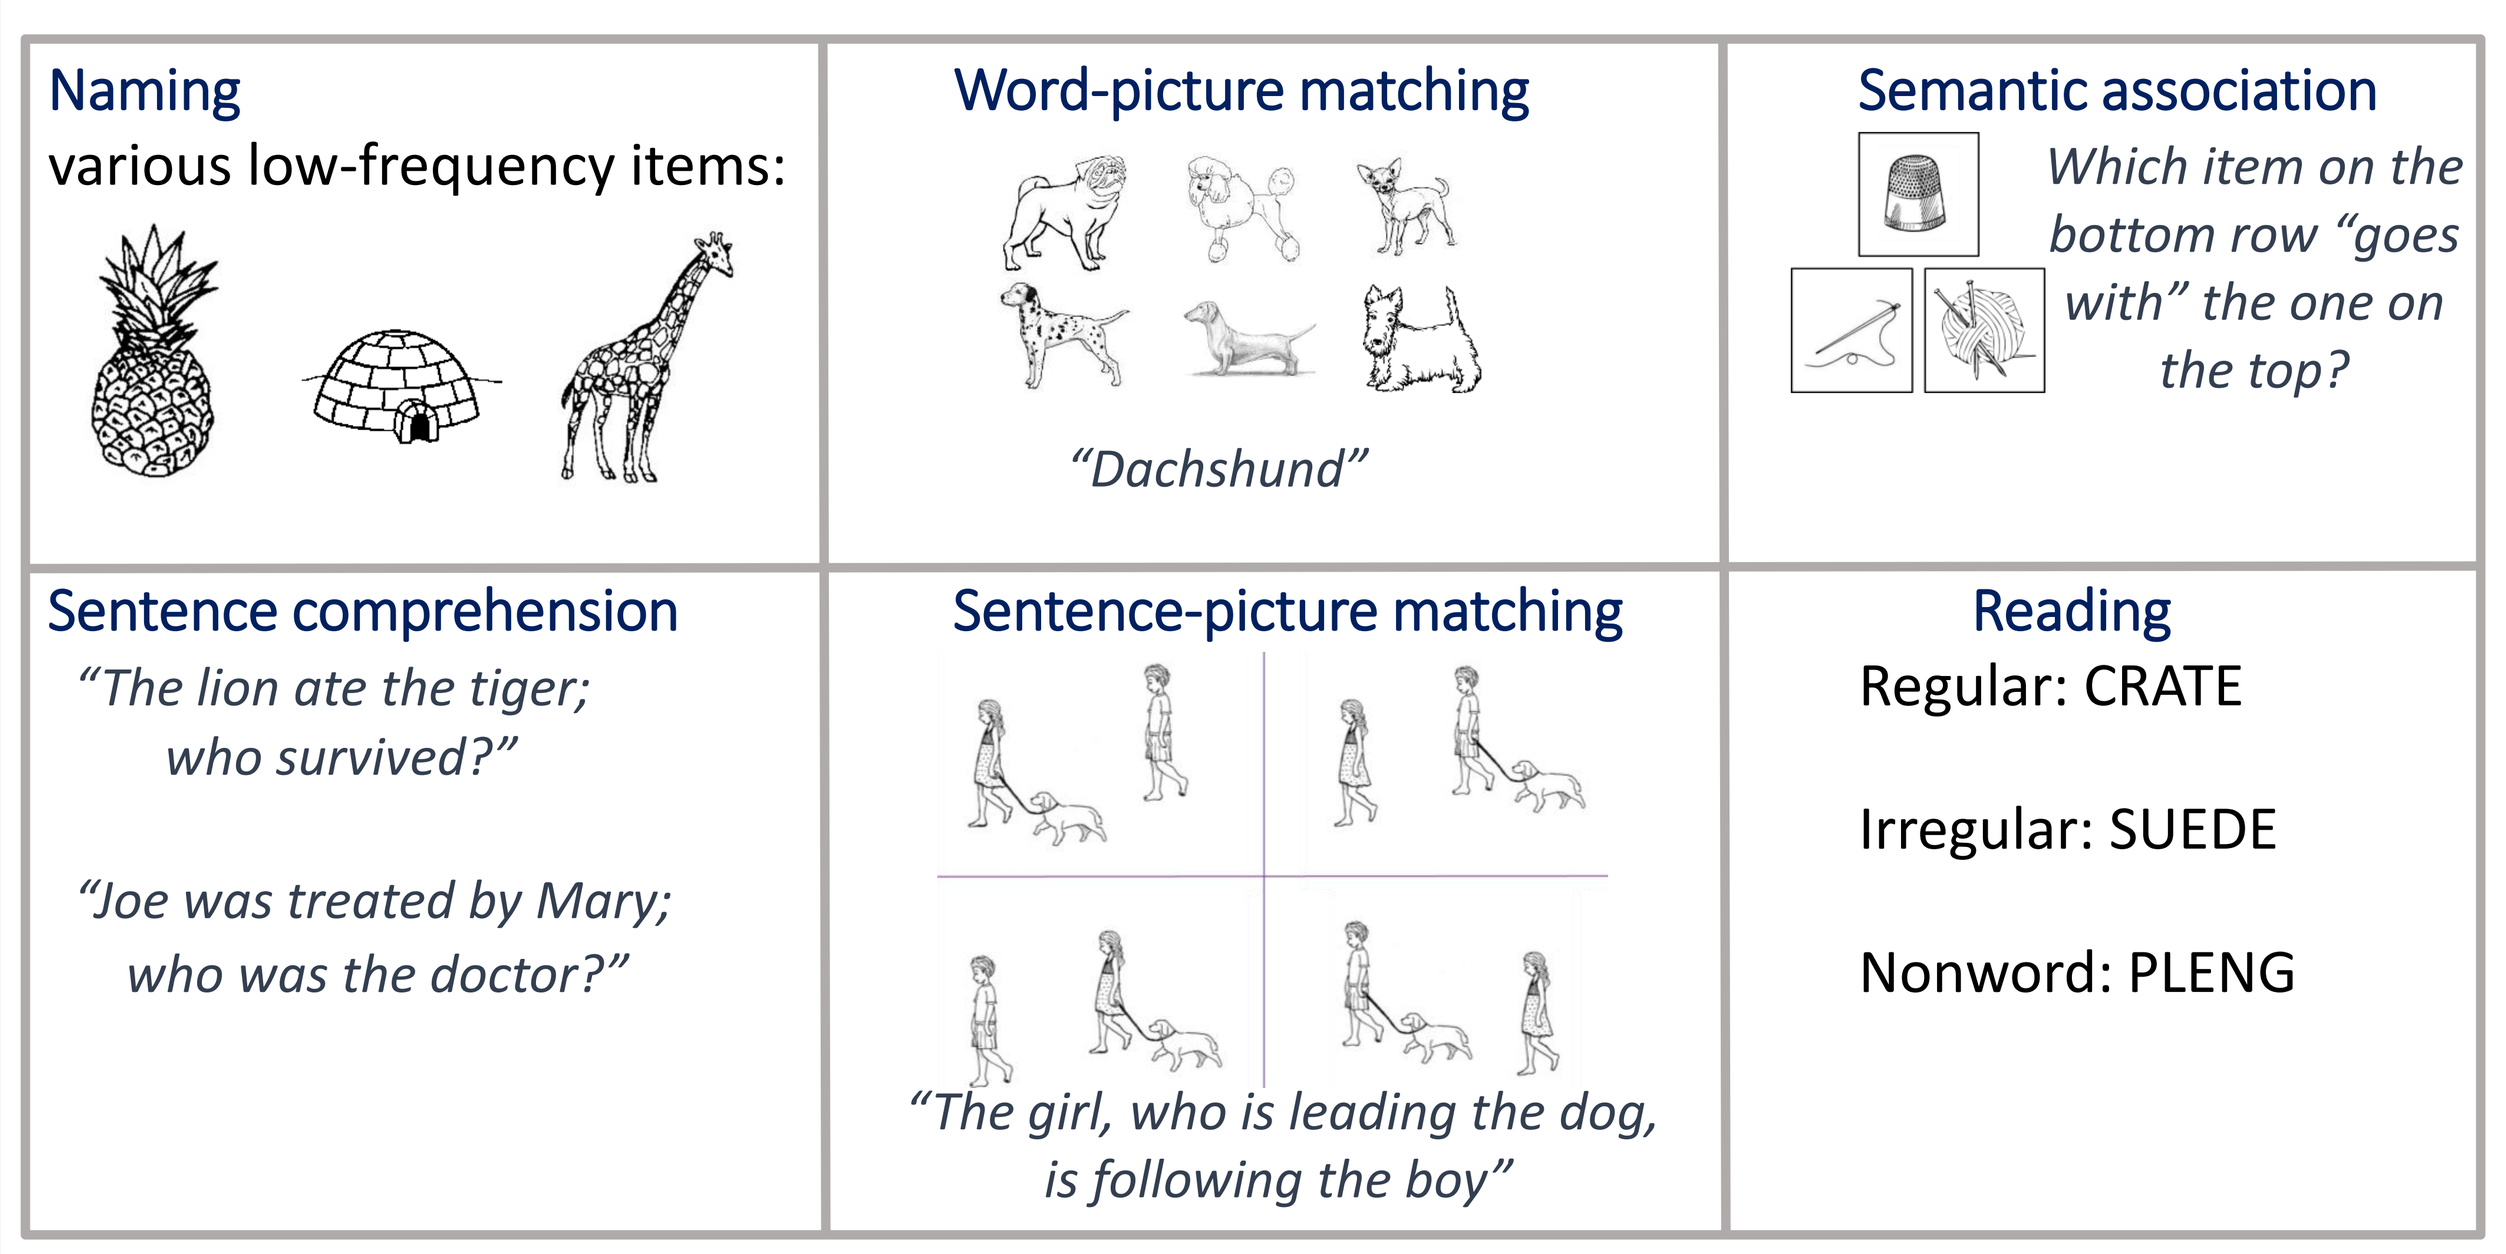
Naming
various low-frequency items:
Word-picture matching
"Dachshund"
Semantic association
Which item on the bottom row "goes with" the one on the top?
Sentence comprehension
"The lion ate the tiger; who survived?"
"Joe was treated by Mary; who was the doctor?"
Sentence-picture matching
"The girl, who is leading the dog, is following the boy"
Reading
Regular: CRATE
Irregular: SUEDE
Nonword: PLENG

Statistical analysis

Group differences between **(1)** PPA patients, movement disorder (MD) patients (the PSP and CBS groups, combined), and controls; and **(2)** PSP and CBS patients in performance on tests of semantic knowledge, syntactic ability, repetition, and reading were examined

Error types

Examining the types of errors made on language tasks can provide useful information about the nature of impairment. Incorrect responses by PSP and CBS patients on the naming and reading tasks were categorised according to error types

Results I: Performance on MLSE subtests (Movement disorder v PPA v Control)

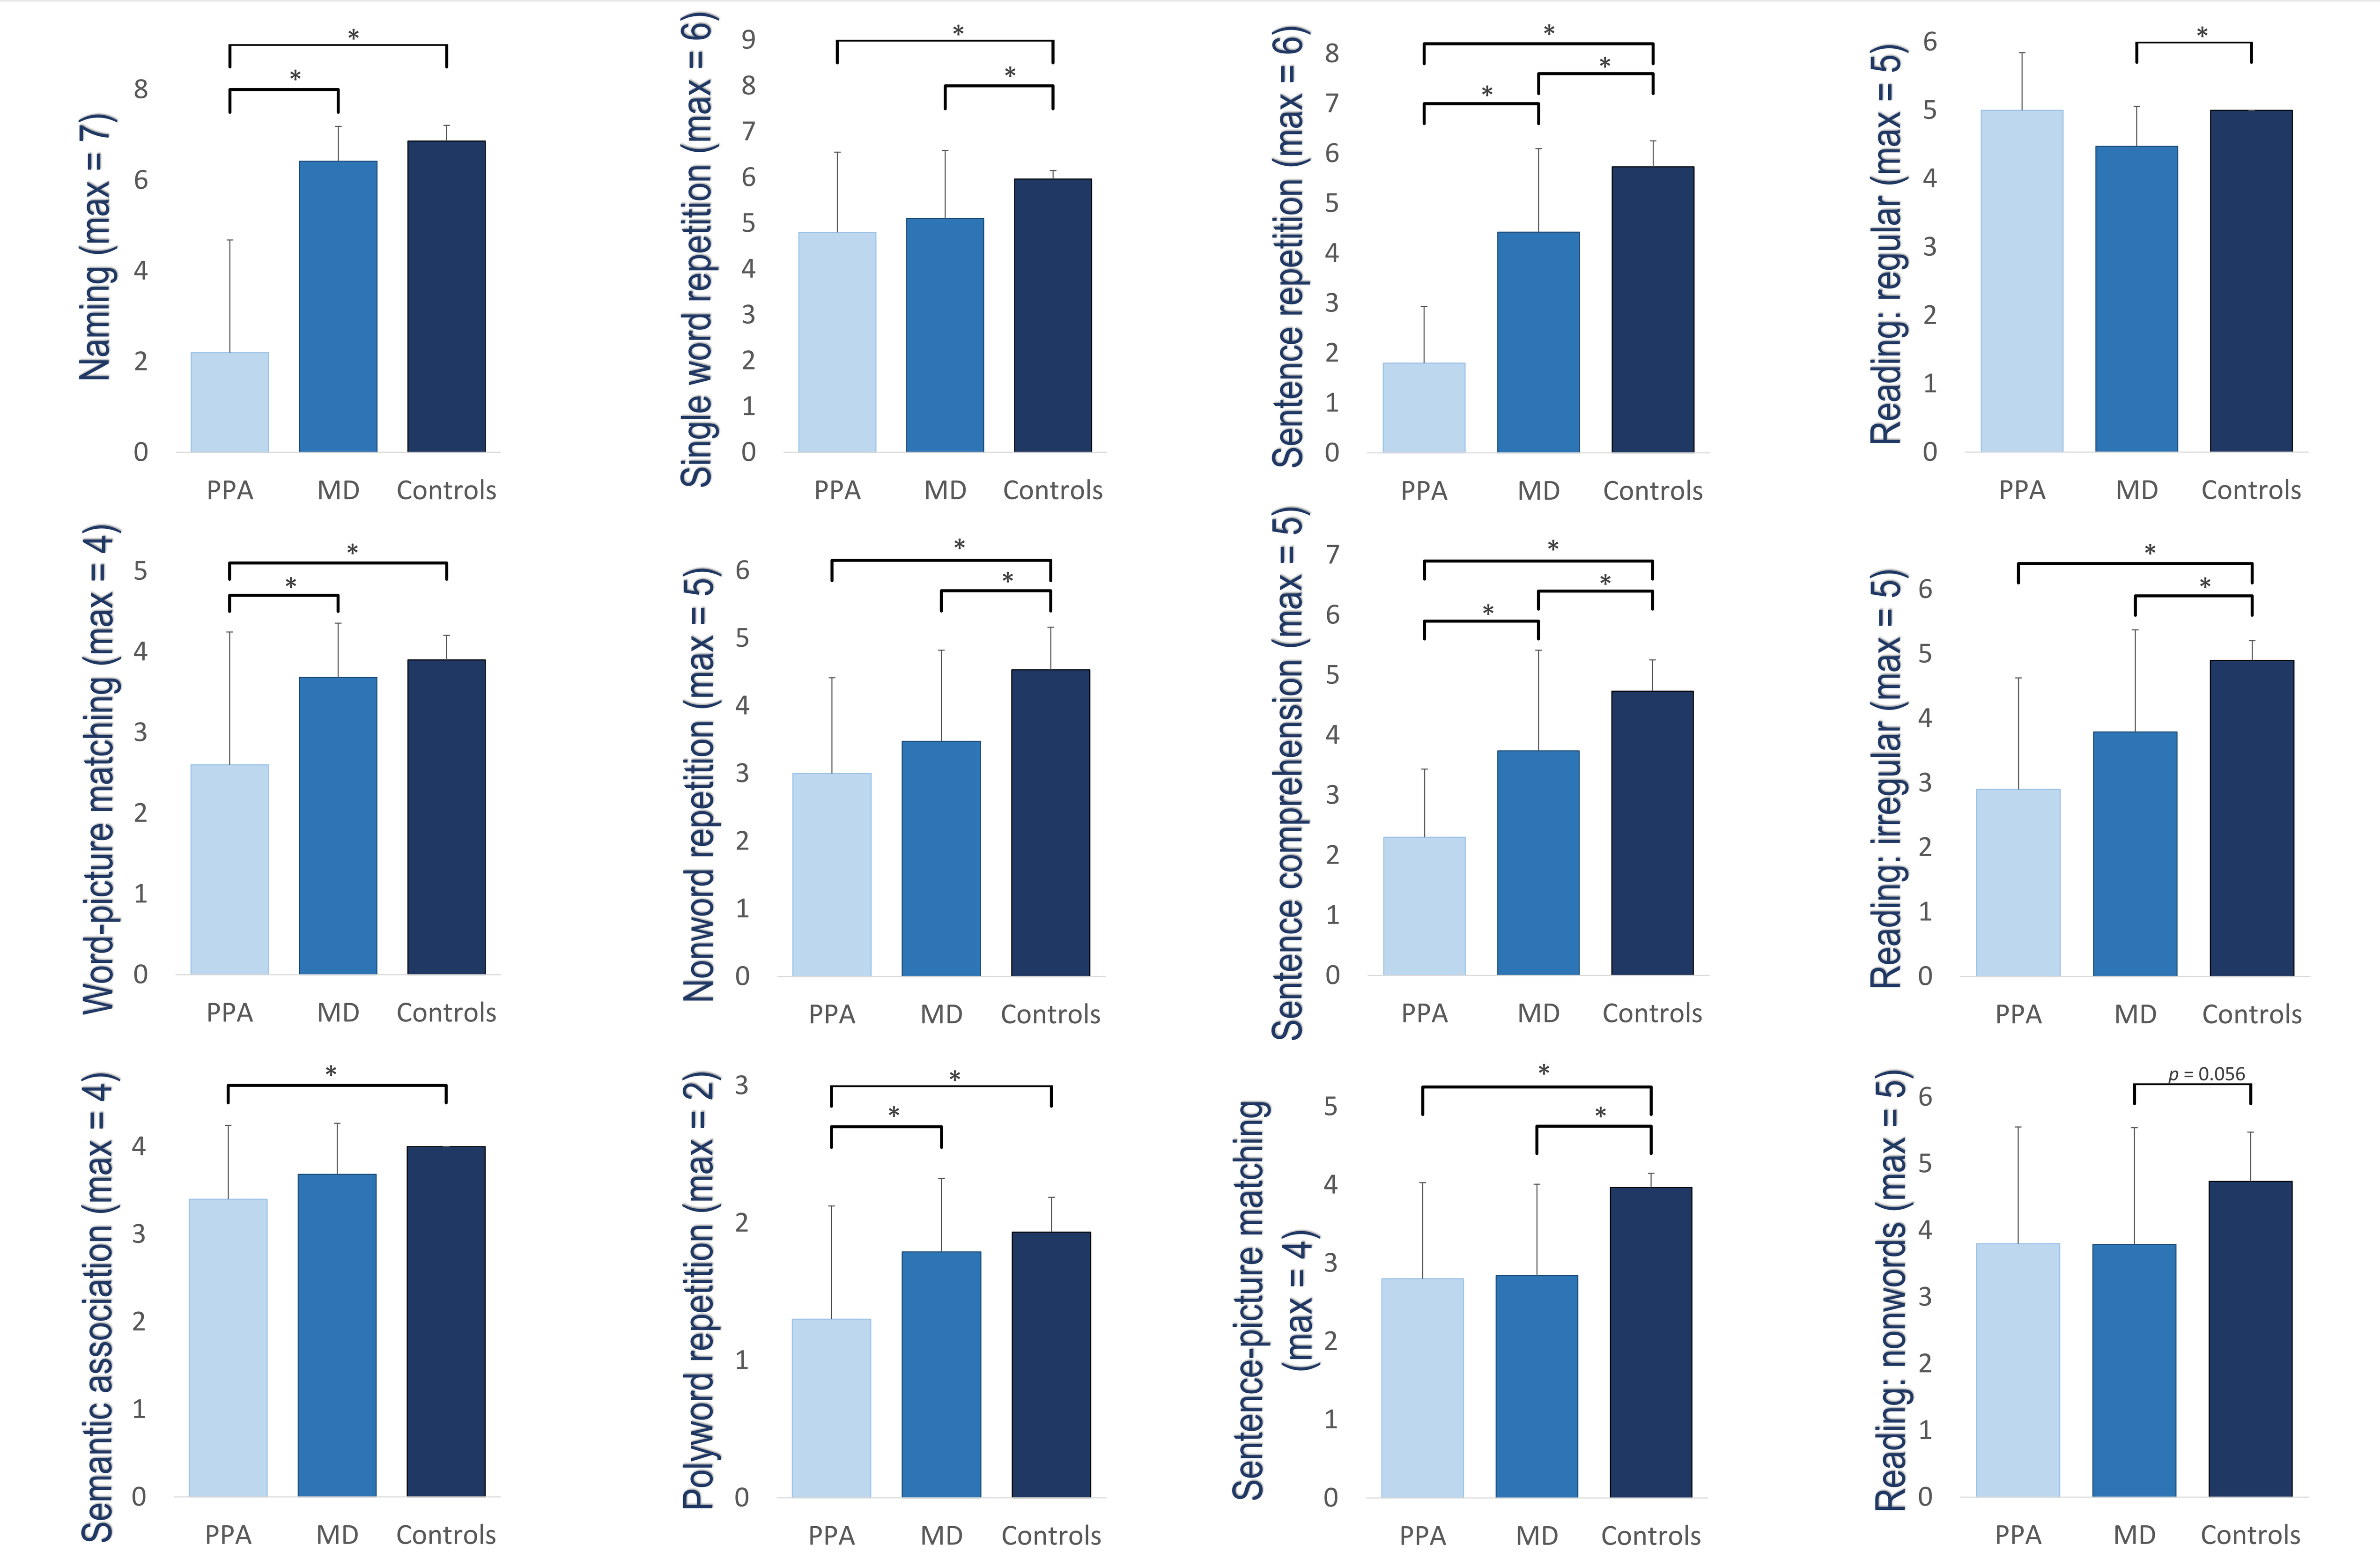
Naming (max = 7)
Single word repetition (max = 6)
Sentence repetition (max = 6)
Reading: regular (max = 5)
Word-picture matching (max = 4)
Nonword repetition (max = 5)
Sentence comprehension (max = 5)
Reading: irregular (max = 5)
Semantic association (max = 4)
Polyword repetition (max = 2)
Sentence-picture matching (max = 4)
Reading: nonwords (max = 5)
p = 0.056
PPA
MD
Controls

Figure 1. Group differences between movement disorder (MD) patients, primary progressive aphasia (PPA) patients, and healthy controls on subtests of the Mini Linguistic State Examination (MLSE)

Scores are mean (SD) number of correct items per subtest. Results were corrected for multiple comparisons using the Bonferroni correction

Results II: PSP v CBS

Overall scores

Group comparisons revealed a significant difference in performance on reading of regular words, with significantly lower scores for PSP patients (mean score = 4, SD = 1.33) than CBS patients (mean score = 5, SD = 0.00; *P* value = 0.038). There were **no other** significant differences between PSP and CBS patients on MLSE subtests

Error types across groups in naming and reading

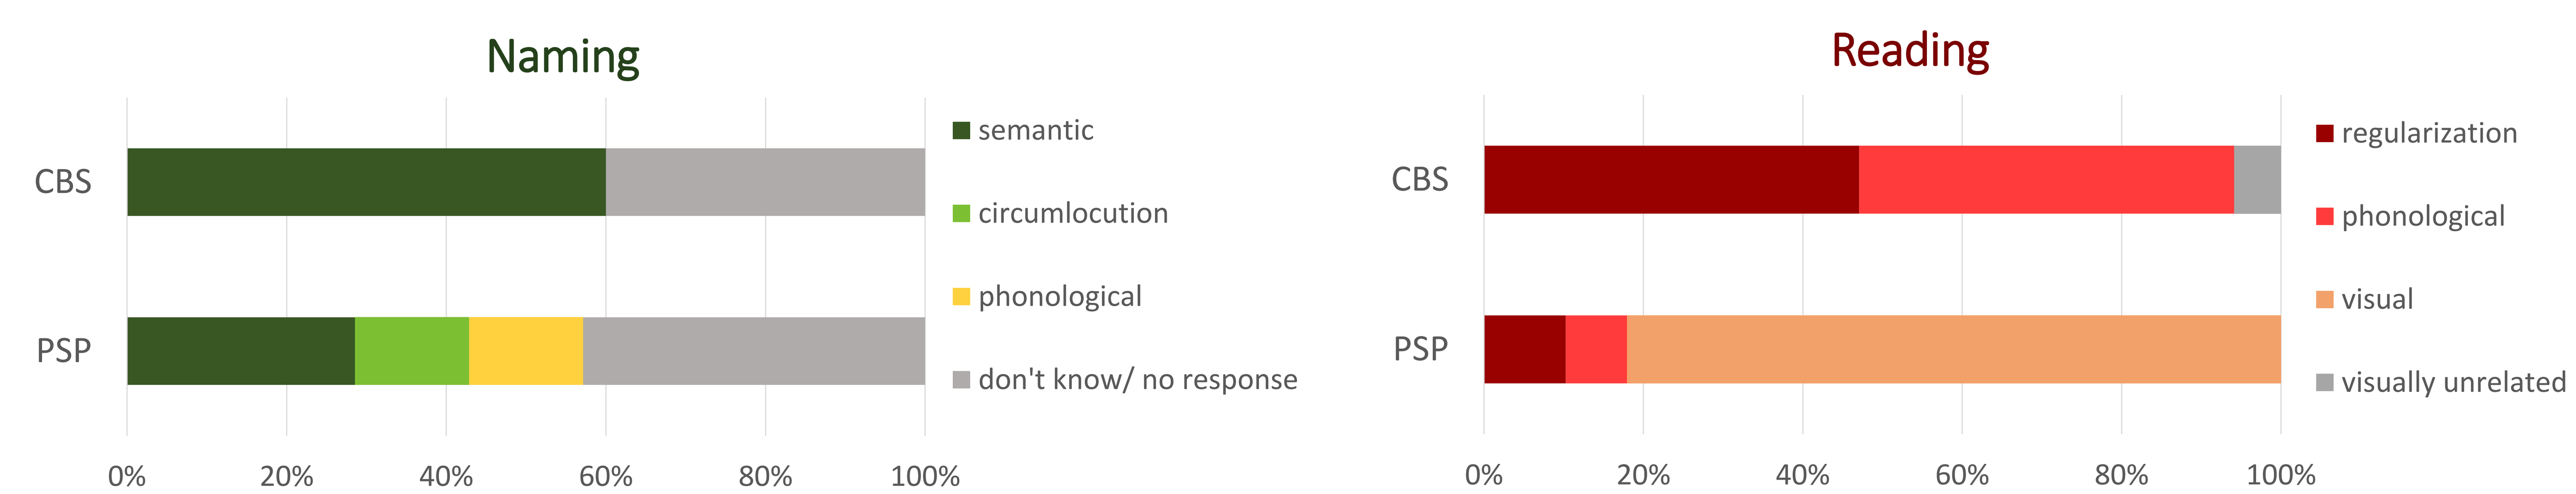
Naming
CBS
PSP
semantic
circumlocution
phonological
don't know/ no response
Reading
regularization
phonological
visual
visually unrelated
0%
20%
40%
60%
80%
100%

Figure 3. Proportion of error types made by PSP and CBS patients (total across group) on naming and reading subtests of the MLSE

Total errors: naming = 5 (CBS) and 7 (PSP); reading = 17 (CBS); and 39 (PSP)

Discussion

- The movement disorder group were significantly impaired on tasks of repetition, reading, and sentence comprehension compared to controls, while PPA patients were significantly more impaired on tests of naming, word-picture matching, polyword repetition, sentence repetition, and sentence comprehension compared to the movement disorder group
- The PSP and CBS groups had similar scores on the majority of the MLSE subtests, suggesting that overall language performance may have limited utility in the differential diagnosis of these two groups
- On the other hand, our preliminary investigation of error types suggests that the two MD groups differ significantly in the nature of their errors on linguistic tasks
- In particular, the majority of reading errors in the PSP group were categorised as visual errors. Visual difficulty is a common feature of PSP and may have a secondary impact on PSP patients' performance on language and other cognitive tests. Indeed, two of the PSP patients in this sample scored 100% on the sentence comprehension task (understanding orally-presented sentences) but only 25% on the sentence-picture matching task. In other words, poor PSP performance on tasks purporting to test syntactic processing, which tend to feature visual stimuli, may in fact be due to impaired visual processing
- Nevertheless, visual difficulty alone cannot account for language impairment in PSP as this group also made a mixture of semantic and phonological errors

Conclusions

- Although PSP and CBS patients may both show mild impairments on tasks of language, there is little evidence that overall language scores will help to differentiate these two disorders
- By contrast, error types – especially on language tasks also requiring visual processing – may aid the differential diagnosis of PSP and CBS

Contact

Dr Katie A. Peterson, Department of Clinical Neurosciences, University of Cambridge. **Email:** kap45@medschl.cam.ac.uk

MLSE study website: www.mlsexam.com

@MLSE_Study